Range Finding Study for Use of *N*-Methyl-*N*-Nitrosourea as a Positive Control Carcinogen in 26-Week Carcinogenicity Study using B6.129-*Trp53*^{tm1Brd} N5 Heterozygous and Wild Type Mice

Taconic
Smart Solutions To Improve Human Health

Kevin Keane,[1] Mark Cartwright,[1] Teresa A. Liberati,[2] Jennifer Criley,[2] Megan M. MacBride[2]

[1]Schering-Plough Research Institute, 144 Route 94, Lafayette, NJ 07848; [2]Taconic Farms, 273 Hover Avenue, Germantown NY 12526

Introduction

Transgenic carcinogenicity testing models represent a valuable alternative accepted by regulatory authorities to the standard 2-year bioassay. The B6.129-*Trp53*^{tm1Brd} N5 heterozygous knockout model (p53$^{+/-}$) is one of the transgenic models approved for use in 26-week carcinogenicity assays. The p53$^{+/-}$ model is used for evaluation of compounds that are suspected to be genotoxic carcinogens. *p*-Cresidine is currently the most commonly used positive control in p53$^{+/-}$ carcinogenicity studies. However, *p*-cresidine poses significant occupational health concerns, and the daily gavage administration necessary is inconvenient and requires additional resources. **The problems inherent in use of *p*-cresidine have spurred the search for a better positive control compound.**

N-Methyl-*N*-nitrosourea (MNU) is the positive control compound of choice for use with the rasH2 short-term carcinogenicity assay. MNU is a much safer compound that provides an adequate positive control with rasH2 mice with just a single dose, by either ip or gavage. The convenience and safety associated with use of MNU prompted this study to assess use of MNU as a positive control in 26-week carcinogenicity studies with p53$^{+/-}$ mice. A single oral dose of MNU is generally well tolerated and results in an increased incidence in neoplasms and proliferative lesions 13 weeks after treatment. **The objective of this study was to assess the appropriate dosing for use of a single oral gavage dose of MNU in B6.129-*Trp53*^{tm1Brd} N5 heterozygous mice (p53$^{+/-}$) and B6.129-*Trp53*^{tm1Brd} N5 wild type mice (wild type or p53$^{+/+}$).**

Method

▪ B6.129-*Trp53*^{tm1Brd} N5 heterozygous and wild type mice were bred at Taconic Farms, Germantown, NY. Animals were 7-8 weeks old at the start of study and were maintained for 13 or 26 weeks.

▪ Mice were group housed during acclimation, dosing and until approximately 24-48 hours post dosing. Once dosed the mice were maintained in disposable caging for approx. 48 hours. Mice were then individually housed in solid bottom microisolator cages with filtered tops and contact bedding for the duration of the study.

▪ *N*-Methyl-*N*-nitrosourea (MNU) was obtained from Sigma Chemical Co., St. Louis, MO. MNU was prepared in 25 mM disodium citrate buffered saline at pH 4.5, with dosages of 0 (vehicle control), 60 and 90 mg/kg used. Dosing solutions were prepared and used within 5 hours on the day of dosing, and analytical analysis was performed on all dosing solutions.

▪ A single oral gavage dose of MNU or vehicle control was administered.

Figure 1: Study design

Group	Total Dose (mg/kg)	Dose Volume (mL/kg)	Dose Conc. (mg/mL)	No. of Males	No. of Females
1[a] Vehicle Control	0	10	0	15/15	15/15
2[a] MNU	60	10	6	15/15	15/15
3[a] MNU	90	10	9	15/15	15/15
4[b] Vehicle Control	0	10	0	15/15	15/15
5[b] MNU	60	10	6	15/15	15/15
6[b] MNU	90	10	9	15/15	15/15

a – p53$^{+/-}$ (heterozygous)
b – p53$^{+/+}$ (wild type)
c – Number of mice at interim necropsy/number of mice at terminal necropsy

Results

Food consumption: There were no significant differences in food consumption between the groups through Week 13 excepting a decrease for both sexes of the 90 mg/kg dosage group for the p53$^{+/-}$ animals. After Week 13, for p53$^{+/-}$ males, decreased food consumption occurred at both 60 and 90 mg/kg as compared to the other groups. In the females, both 90 mg/kg dosage groups, regardless of genotype, had decreased food consumption. The mortality seen in these high dose groups resulted in a smaller number of animals for analysis at later time points.

Figure 2: Food Consumption: A) Males, B) Females

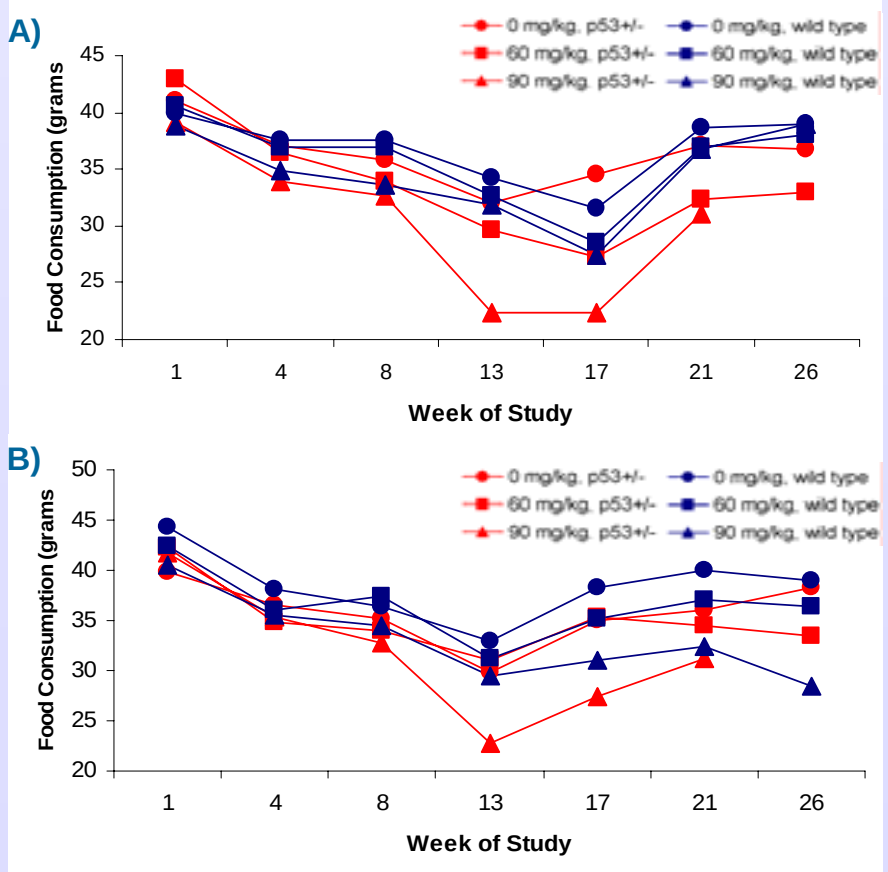

A)
B)
Food Consumption (grams
Week of Study
0 mg/kg, p53+/-
60 mg/kg, p53+/-
90 mg/kg, p53+/-
0 mg/kg, wild type
60 mg/kg, wild type
90 mg/kg, wild type

Body weight: Male mice in all dosage groups except the 90 mg/kg p53$^{+/-}$ mice gained weight in a similar manner throughout the 26 week study. The p53$^{+/-}$ 90 mg/kg dosage group plateaued in body weight at approx. 8 weeks post dosing. Female mice in all dosage groups except the two 90 mg/kg groups also gained weight in a similar manner throughout the study period. Both female 90 mg/kg dosage groups demonstrated a plateau in body weight beginning at approximately 13 weeks post dosing, though the body weights for the few surviving female mice in the 90 mg/kg p53$^{+/-}$ group increased again after week 17. This represents just 2 mice left at week 21 and 1 mouse left at week 26 for that group. Survival was higher at those time points in the wild type females in the 90 mg/kg dose group.

Figure 3: Male Body Weights

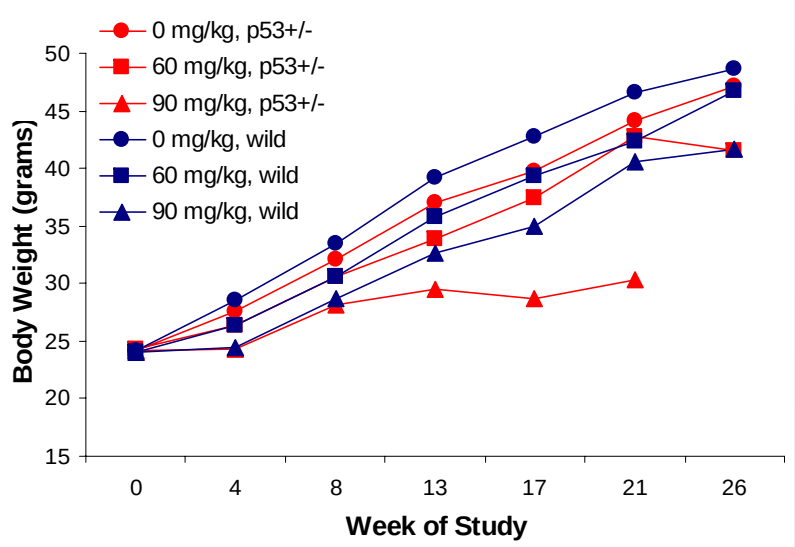

Body Weight (grams)
Week of Study
0 mg/kg, p53+/-
60 mg/kg, p53+/-
90 mg/kg, p53+/-
0 mg/kg, wild
60 mg/kg, wild
90 mg/kg, wild

Figure 4: Female Body Weights

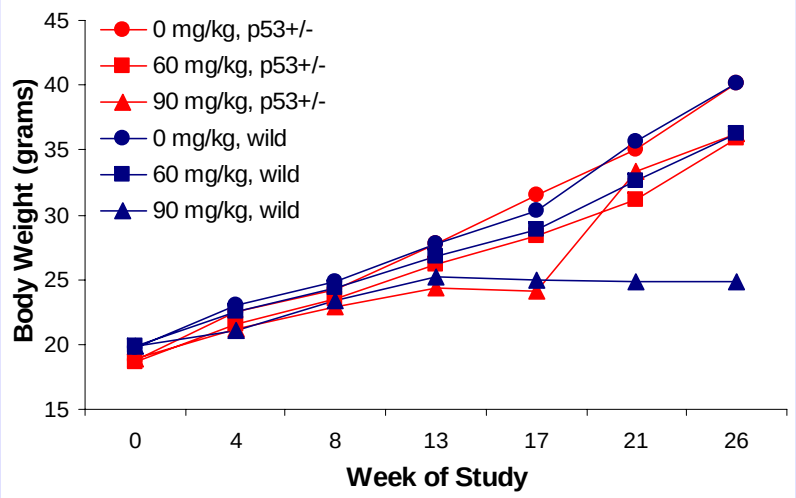

Body Weight (grams)
Week of Study
0 mg/kg, p53+/-
60 mg/kg, p53+/-
90 mg/kg, p53+/-
0 mg/kg, wild
60 mg/kg, wild
90 mg/kg, wild

Survival: The rate of decedents was increased in the p53$^{+/-}$, MNU-dose groups compared to the wild type MNU-dose groups. The survival rates between the p53$^{+/-}$ and wild type control groups were comparable. Deaths occurred beginning at approximately 10 weeks post-dosing; however, the majority of the deaths occurred after 14 weeks on study. Only six animals were found dead in the Group 1 or 4 vehicle control groups.

Figure 5: Male Survival Curve

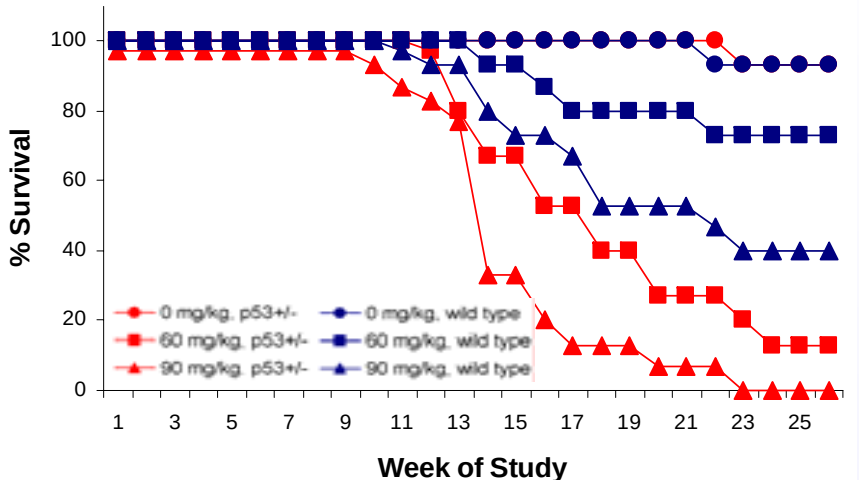

% Survival
Week of Study
0 mg/kg, p53+/-
60 mg/kg, p53+/-
90 mg/kg, p53+/-
0 mg/kg, wild type
60 mg/kg, wild type
90 mg/kg, wild type

Figure 6: Female Survival Curve

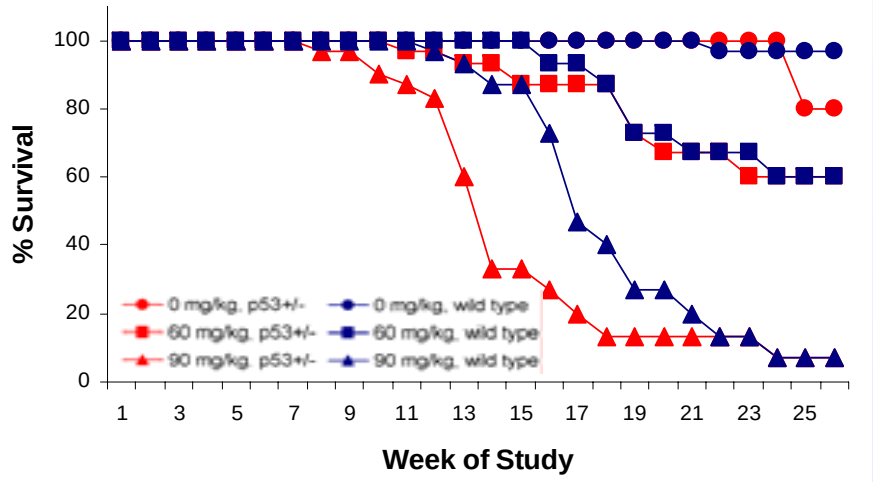

% Survival
Week of Study
0 mg/kg, p53+/-
60 mg/kg, p53+/-
90 mg/kg, p53+/-
0 mg/kg, wild type
60 mg/kg, wild type
90 mg/kg, wild type

Conclusions

Test article-related mortality was observed in all MNU dose groups. **The MNU-related mortality was higher in the p53$^{+/-}$ groups compared to the wild type groups.** Based on the mortality, the 90 mg/kg dosage of MNU exceeded the maximum tolerated dose. At the 60 mg/kg dosage survival to 26 weeks appears acceptable. In comparison, daily treatment of p53$^{+/-}$ mice with 400 mg/kg *p*-cresidine by oral gavage resulted in 86% survival in males and 80% survival in females over 26 weeks.[1] The bladder tumors induced by *p*-cresidine treatment are likely not as fatal as MNU-induced tumors, meaning that an appropriate positive control MNU dosage may result in higher mortality compared to *p*-cresidine control groups.

Previous work involving MNU treatment (30 or 120 ppm in drinking water) of p53$^{+/-}$ and wild type mice demonstrated a genotype-dependent effect, with significantly increased mortality in p53$^{+/-}$ versus wild type mice. This mortality was attributed to MNU-induced lymphomas, leukemias or sarcomas.[2] For the purpose of this dose range-finding study, mortality may thus be used as a surrogate marker of tumorigenesis. **In males, a genotype-dependent increase in mortality was seen at both MNU dosages, whereas in females only the 90 mg/kg MNU dose resulted in decreased latency in mortality.**

Acceptance of a single oral gavage dose of MNU as a suitable positive control in place of the currently used *p*-cresidine would make performance of 26-week carcinogenicity assays with p53+/- mice easier and safer. **MNU should be useful as a positive control for short term carcinogenicity assays using p53$^{+/-}$ transgenic mice, although further refinement of an acceptable dosage is warranted.** Other refinements could consist of definition of an interim necropsy time point for the positive control group or study conduct involving appropriate handling processes in order to collect data from the high percentage of found dead or early terminated mice.

Acknowledgements - This study was supported through a grant from the ILSI Health and Environmental Sciences Institute.

1. Stoll, R.E.; Blanchard, K.T.; Stoltz, J.H.; Majeska, J.B.; Furst, S.; Lilly, P.D.; Mennear, J.H. *Toxicol. Sci.* **2006**, *90, 440-450.*
2. Yamamoto, M.; Tsukamoto, T.; Sakai, H.; Shirai, N.; Ohgaki, H.; Furihata, C.; Donehower, L.A.; Yoshida, K.; Tatematsu, M. *Carcinogenesis*, **2000**, *21*, 1891-1897.